HydroFix™ – Posterior Laminectomy Sheep Study

Paul Jeffords MD[1], Guilhem Denoziere MS[2], Rebeccah Brown PhD[2]

1. Resurgens Spine Center, 5671 Peachtree Dunwoody Rd # 900 Atlanta, GA 30342-5022 2. MiMedx Group, 811 Livingston Ct SE # B, Marietta, GA 30067-8958

Introduction

The MiMedx HydroFix™ Spine Shield is a unique protective "hydrogel" surgical barrier created from a hydrated polyvinyl alcohol (PVA) substrate. The thin, flexible Spine Shield is indicated for use as a cover for the spine, including contact with the Central Nervous System and Central Circulatory System, to provide a plane of dissection during revision surgery. In the event of needed revision surgery, the device protects underlying structures, during the surgical approach and is proposed to better permit localization of the prior surgical site through improved tissue plane dissection as well. The current study looks at the long term effect of placement of the HydroFix™ Spine Shield in sheep, to confirm the devices are inert, nonadherent, biocompatible, and durable while maintaining long term protection of the spine and facilitating tissue dissection in revision surgery.

Materials and Methods

The devices were implanted into 8 sheep following a posterior laminectomy approach by a board certified orthopedic surgeon using standard instrumentation and technical procedures. 3 sheep each were then evaluated with an explant procedure at 30 and 90 days, and 2 sheep at 180 days to determine key properties of the device. At each surgery, three surgeons were present to independently evaluate the gross anatomical effectiveness of the product and to score the ability to separate the overlying structures from the previous surgical site. In addition, extensive sampling was undertaken to evaluate micropathological environment and success of the shields.

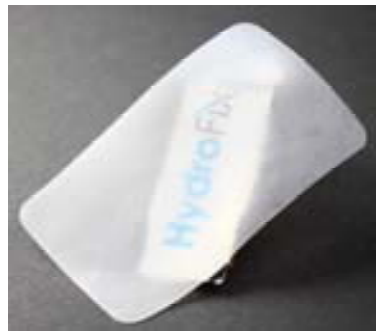
HydroFix

Figure 1. HydroFix™ Spine Shield

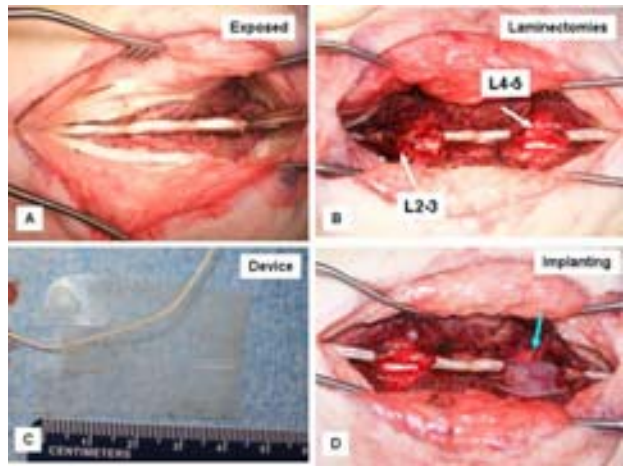
Exposed
Laminectomies
L4-5
L2-3
Device
Implanting
A
B
C
D

Figure 2. Implantation Technique

Results

Blood collected on the day of the implantation surgery, at mid-way through the in-life phase and prior to euthanasia showed normal values for overall average blood hematology and chemistry results.

Neurological examinations conducted on all animals prior to implant and at multiple time points during the study did not show any abnormal findings.

Animals in the 180 day group underwent MRI scans at 102 days post implant. No depression of the spinal cord was observed at the test site in one sheep (Figure 3a) and a slight depression of the spinal cord was observed in the second sheep (Figure 3b), with however no abnormal neurological finding.

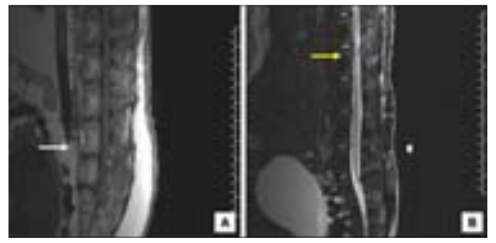
A
B

Figure 3: MRI Scans

The plane of dissection scores at the 30, 90 day and 180 day time points were significantly lower at the device site than the control site (P<0.003).

Score	Description
1	**Separates with no adhesion**- applicable tissues can be safely separated from the study site without the use of surgical tools
2	**Easily detachable**- applicable tissues can be safely separated from the study site with minimal use of blunt surgical tools to overcome light adhesion
3	**Dissection required**- applicable tissues can be safely separated from the study site while using blunt surgical tools to overcome moderate adhesion
4	**Sharp dissection required**-applicable tissues cannot be separated from the study site without risk of damage as the use of sharp surgical tools is required to overcome tenacious adhesion

Metric	Range	30-Day Average	90-Day Average	180-Day Average
Scores for revision with HydroFix Spine Shield	1 - 2	1.0	1.3	1.2
Scores for revision without HydroFix Spine Shield	2 - 4	2.6	3.7	3.3

Figure 4: Plane of Dissection Scores

In the 30-day group, the control and treatment laminectomy sites were filled with overlying soft tissue. At the control site, the surgical surface was covered with a thick band of fibrous connective tissue. At the treatment site, a thin band of fibrotic connective tissue bridged the laminectomy gap between bilateral segments of excised bones, and the fibrous connective tissue formed a pocket around the device over the spinal cord. Host responses to tissue disruption included mild lymphocytic infiltration.

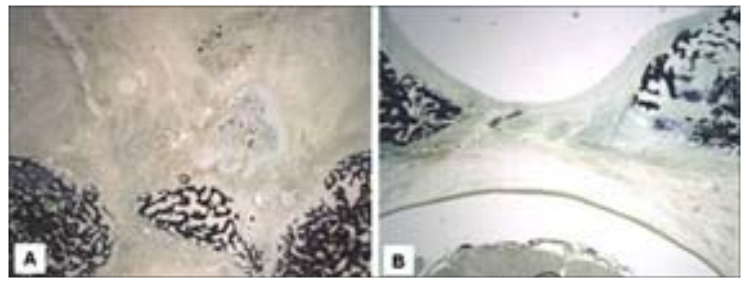
A
B

Figure 5: 30 Day Histology (A. Control
B. Treatment)

In the 90 day group, at the treatment sites, a thin band of fibrotic connective tissue bridged the laminectomy gap and formed a pocket around the device over the spinal cord Also in the 90-day group, more surface calcifications were found at both laminectomy sites compared to the 30-day group. There was more fibrosis in the laminectomy gap at the control sites of the 90-day group than in the control sites of the 30-day group. The inflammatory response was regressive on both sites at 90 days when compared to the 30-day group.

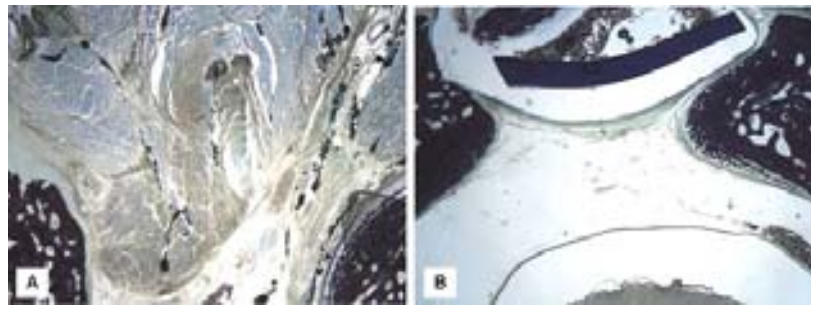
A
B

Figure 6: 90 Day Histology (A. Control, B. Treatment)

In the 180-day group, a band of connective tissue bridged the laminectomy gap over the spinal cord in one animal. In the other animal, abundant angiogenesis was found in the connective tissue and the laminectomy area was fused. The progressive calcifications observed during the study is characteristic of a sheep model undergoing bone defects.

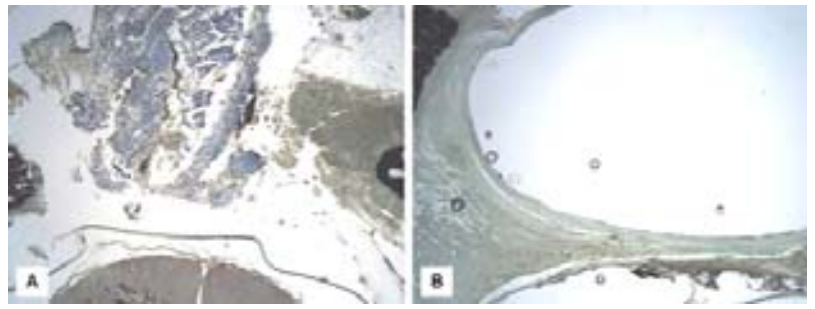
A
B

Figure 7: 180 Day Histology (A. Control , B. Treatment)

Histology was conducted on all devices at three different locations. The strands were non-degenerate and there were no cells, exudates or other tissues associated with the material. Figure 8 shows representative sections of the device at the 90-day time point.

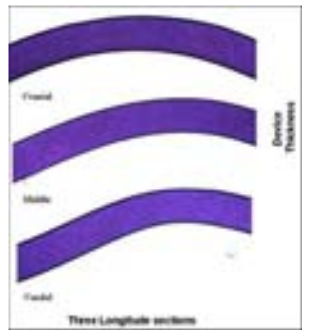
Device Thickness

Figure 8: 90 Day Device Histology

For both groups, the sections taken from visceral organs, brain and spinal cord were normal.

Conclusion

The results support the HydroFix™ Spine Shield device safety and effectiveness in reducing the risk of potential neural tissue damage during posterior vertebral revision surgery by providing a plane of dissection. The current study confirms through placement of the HydroFix™ Spine Shield in sheep that the devices are inert, nonadherent, biocompatible, and durable while maintaining long term protection of the spine and facilitating tissue dissection in revision surgery.

For further information

MiMedx Group, Inc.
811 Livingston Ct. SE, Suite B
Marietta, GA 30067
Toll Free: (866) 477-4219
www.mimedx.com

MiMedx Group 2/11

NOT AVAILABLE IN THE UNITED STATES